Institutional Repository - Research Portal
Dépôt Institutionnel - Portail de la Recherche

researchportal.unamur.be

University of Namur

RESEARCH OUTPUTS / RÉSULTATS DE RECHERCHE

Cardiovascular effect of BCR-ABL TKIs: a meta-analysis and systematic review of arterial and venous occlusive events

Haguet, Hélène; Mullier, François; Chatelain, Christian; Graux, Carlos; Dogné, Jean-Michel; Douxfils, Jonathan

Publication date:
2016

Link to publication

Citation for pulished version (HARVARD):
Haguet, H, Mullier, F, Chatelain, C, Graux, C, Dogné, J-M & Douxfils, J 2016, 'Cardiovascular effect of BCR-ABL TKIs: a meta-analysis and systematic review of arterial and venous occlusive events', 62nd Annual SSC Meeting of the International Society of Thrombosis and Hemostasis, Montpellier, France, 25/05/16 - 28/05/16.

General rights
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.

• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal ?

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

Download date: 25. Apr. 2024

HEM08

Meta-analysis of the risks of arterial and venous occlusive events with new generation BCR-ABL TKIs in patients with chronic myeloid leukemia

Hélène Haguet*[1, 2], Jonathan Douxfils[1], François Mullier[2], Christian Chatelain[1], Carlos Graux[3], Jean-Michel Dogné[1]

[1] University of Namur, Department of pharmacy, Belgium [2]CHU UCL Namur, Hematology laboratory, Belgium [3]CHU UCL Namur, Department of hematology, Belgium

Background

High rate of arterial and venous occlusive events were reported with ponatinib during clinical development[1] and serious cases of arterial occlusive disease were also reported with nilotinib.[2] This led to the evaluation of the vascular safety profile of new generation BCR-ABL TKIs through a meta-analysis that confirmed the increased risk of vascular occlusive events with ponatinib and nilotinib compared with imatinib in chronic myeloid leukaemia (CML). The risk was also with dasatinib.[3] However, distinction between arterial of venous events was not assessed.

Objectives

- To determine the risk of arterial and venous occlusive events in patients with Ph+ CML treated with new generation BCR-ABL TKIs in randomized clinical trials.
- Stratifications by treatment are performed to provide product specific risk assessment.

Methods

Literature search
- Screening of scientific articles (PubMed, Scopus, Cochrane library), congress abstracts (ASH, ASCO, ESMO) and clinical trial register (www.clinicaltrials.gov)
- Selection of all randomized clinical trials comparing new generation TKIs versus imatinib in patients with Ph+ CML.

Data collection
- Study and population characteristics
- Arterial occlusive events
- Venous occlusive events

Statistical analysis
- Random- (REM) and fixed-effect models (FEM) have been used to analyze the risk of arterial occlusive events and venous occlusive events respectively.
- Effect size measure: odds ratio computing using Peto method
- Heterogeneity assessment: Cochran's Q statistic and I^2 value.
- One-way sensitivity analysis was performed to assess the robustness

Results

Figure 1. PRISMA (Preferred Reporting Items for Systematic review and Meta-Analysis) flow diagram of study selection

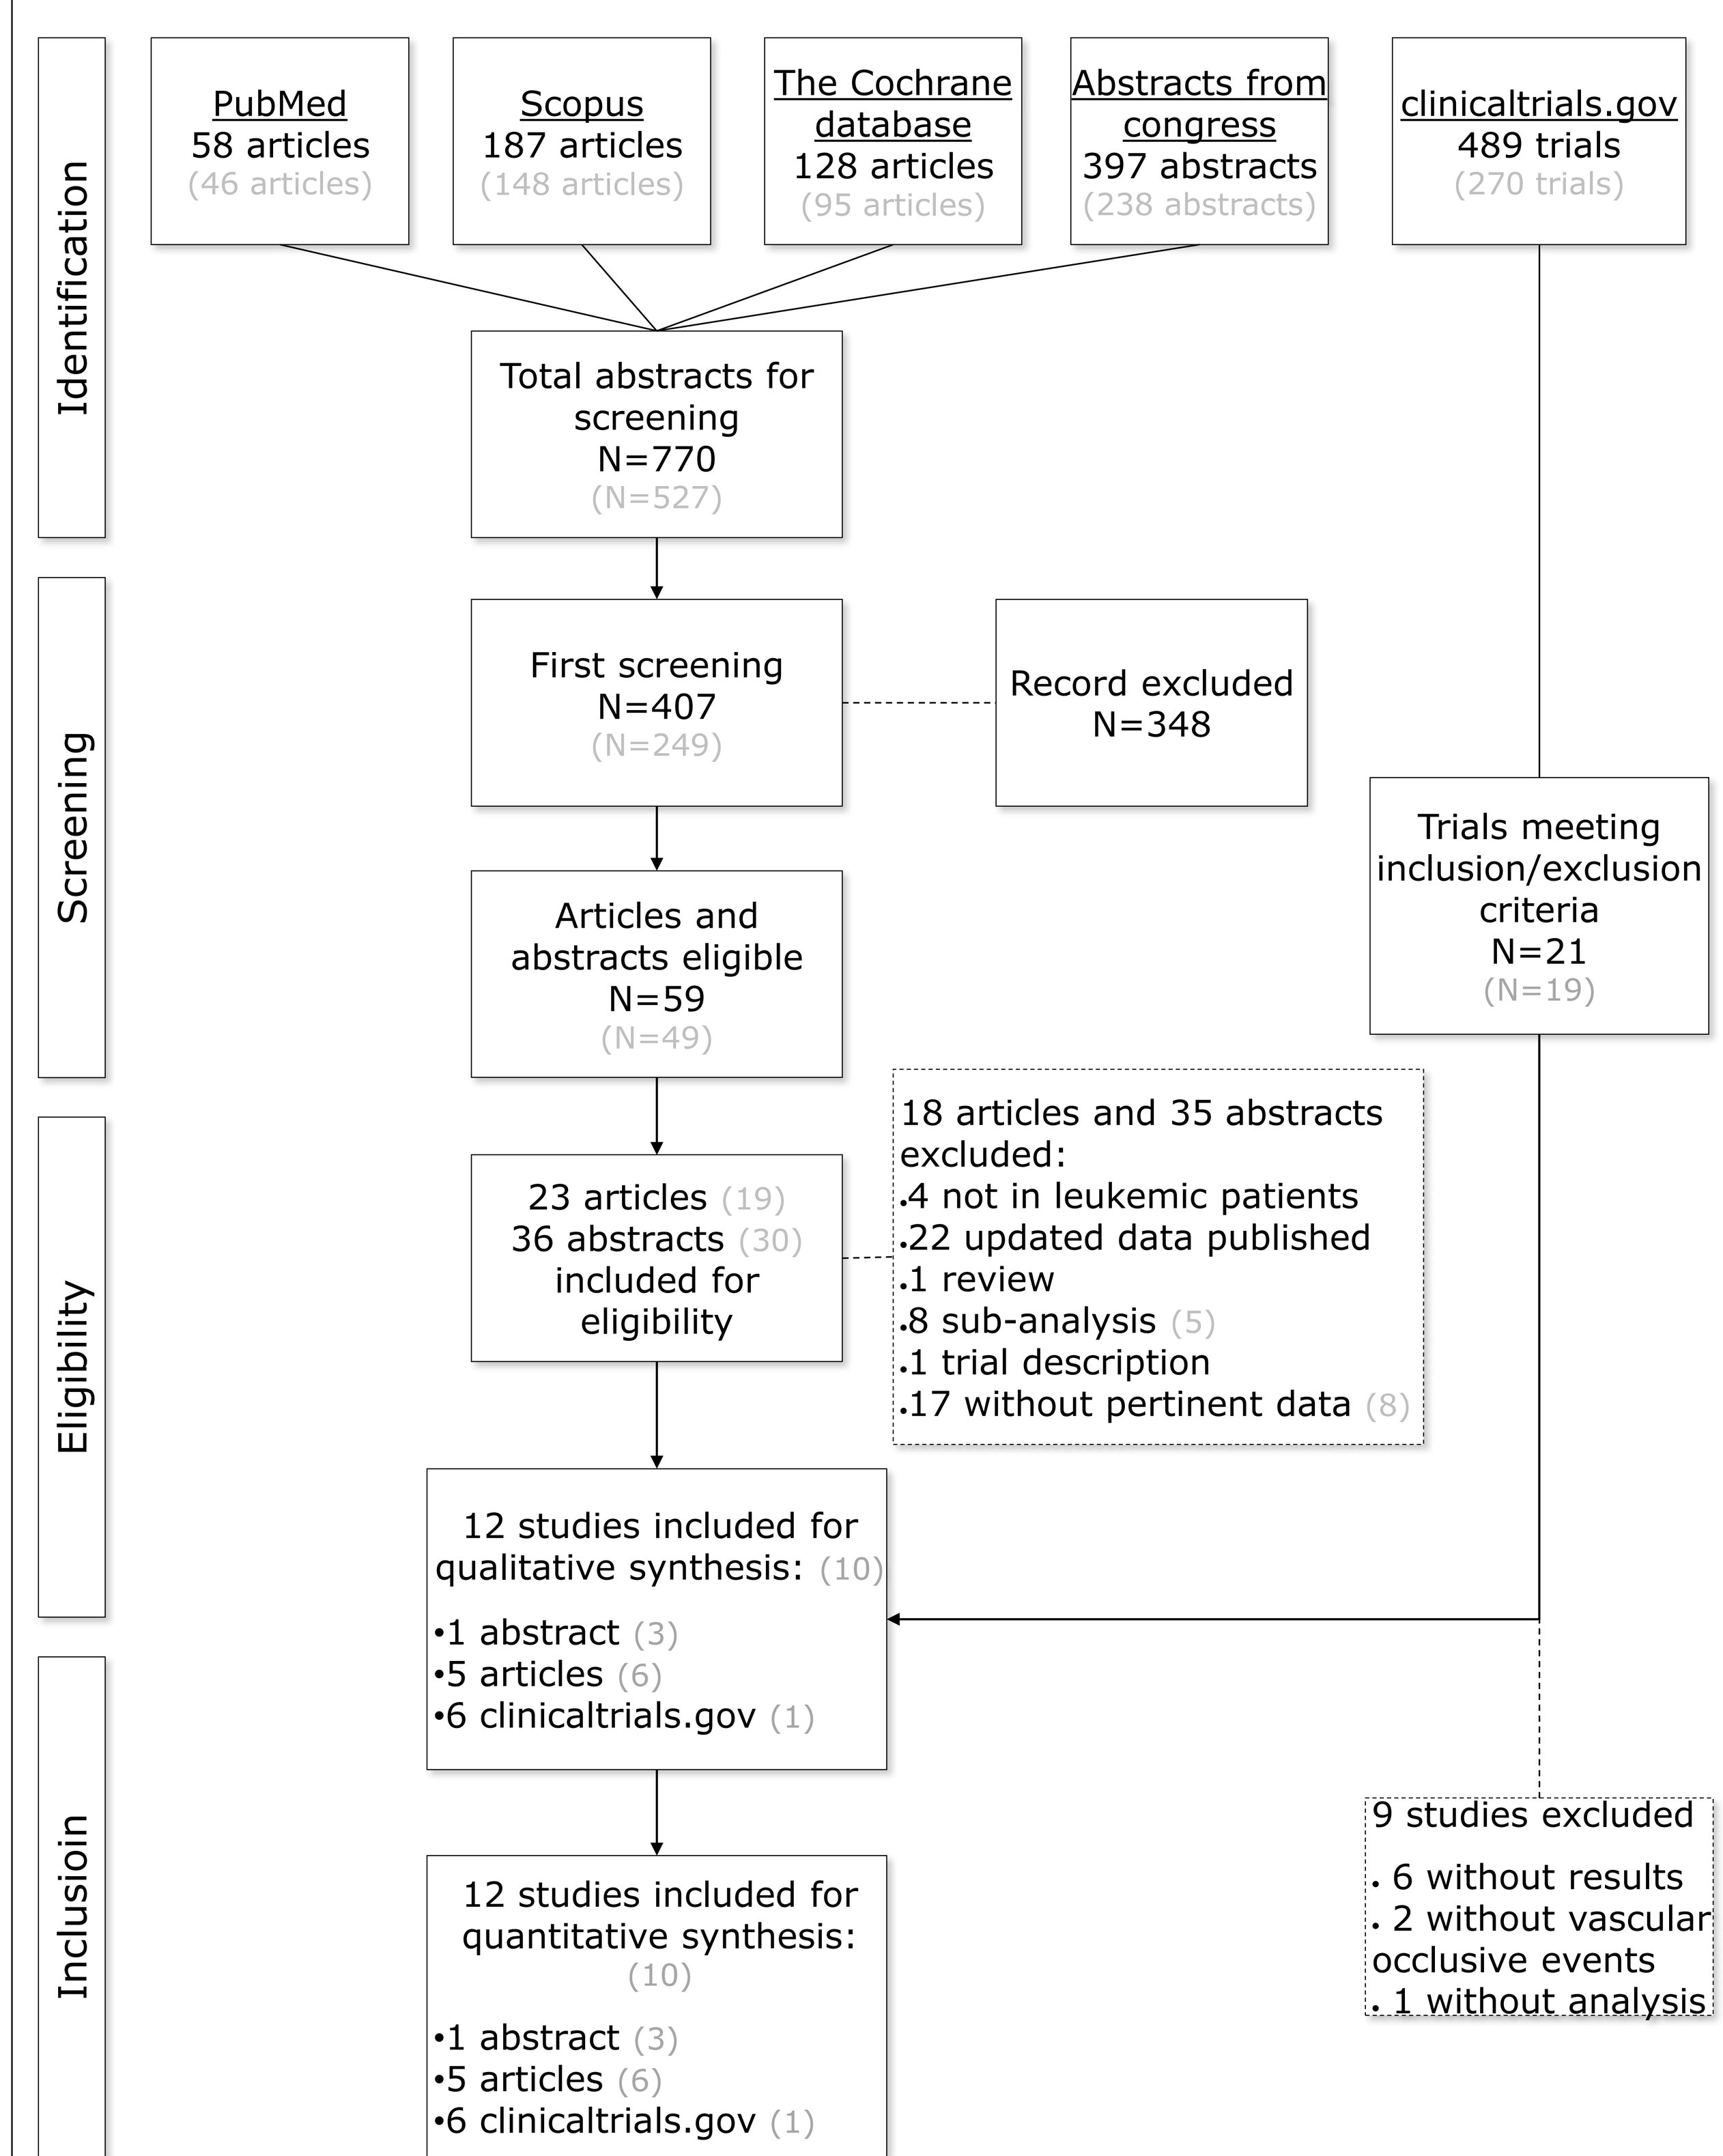
Identification
Screening
Eligibility
Inclusioin
PubMed 58 articles (46 articles)
Scopus 187 articles (148 articles)
The Cochrane database 128 articles (95 articles)
Abstracts from congress 397 abstracts (238 abstracts)
clinicaltrials.gov 489 trials (270 trials)
Total abstracts for screening N=770 (N=527)
First screening N=407 (N=249)
Record excluded N=348
Articles and abstracts eligible N=59 (N=49)
Trials meeting inclusion/exclusion criteria N=21 (N=19)
23 articles (19) 36 abstracts (30) included for eligibility
18 articles and 35 abstracts excluded:
.4 not in leukemic patients
.22 updated data published
.1 review
.8 sub-analysis (5)
.1 trial description
.17 without pertinent data (8)
12 studies included for qualitative synthesis: (10)
•1 abstract (3)
•5 articles (6)
•6 clinicaltrials.gov (1)
9 studies excluded
. 6 without results
. 2 without vascular occlusive events
. 1 without analysis
12 studies included for quantitative synthesis: (10)
•1 abstract (3)
•5 articles (6)
•6 clinicaltrials.gov (1)

- Twelve studies fulfilled the established criteria and were included in the meta-analysis.

Table 1: Absolute risk of arterial and venous occlusive events in patients with CML.

Treatments	Venous occlusive events		Arterial occlusive events	
	New generation TKIs	Imatinib	New generation TKIs	Imatinib
Bosutinib	0/248 (0.00)	0/251 (0.00)	3/248 (1.21)	1/251 (0.40)
Nilotinib	4/886 (0.45)	0/608 (0.00)	69/886 (7.79)	7/608 (1.15)
Dasatinib	8/929 (0.86)	3/873 (0.34)	16/929 (1.72)	4/873 (0.46)
Ponatinib	1/154 (0.65)	0/152 (0.00)	11/154 (7.14)	3/152 (1.97)
Overall	**13/2217 (0.59)**	**3/1884 (0.16)**	**99/2217 (4.47)**	**15/1884 (0.80)**

- Overall, 4.47% (99/2,217) of patients developed arterial occlusive events with new generation BCR-ABL TKIs compared with 0.80% (15/1,884) with imatinib (REM OR_{PETO}: 3.46; 95%CI: 2.35 to 5.10).
- Venous occlusive events occurred in only 0.86% (13/2,217) of patients treated with new generation TKIs and in 0.16% (3/1,884) of imatinib-treated patients.

Figure 2: Forest plots of arterial and venous occlusive events in patients with Ph+ CML treated with new generation TKIs versus imatinib.

Arterial occlusive events

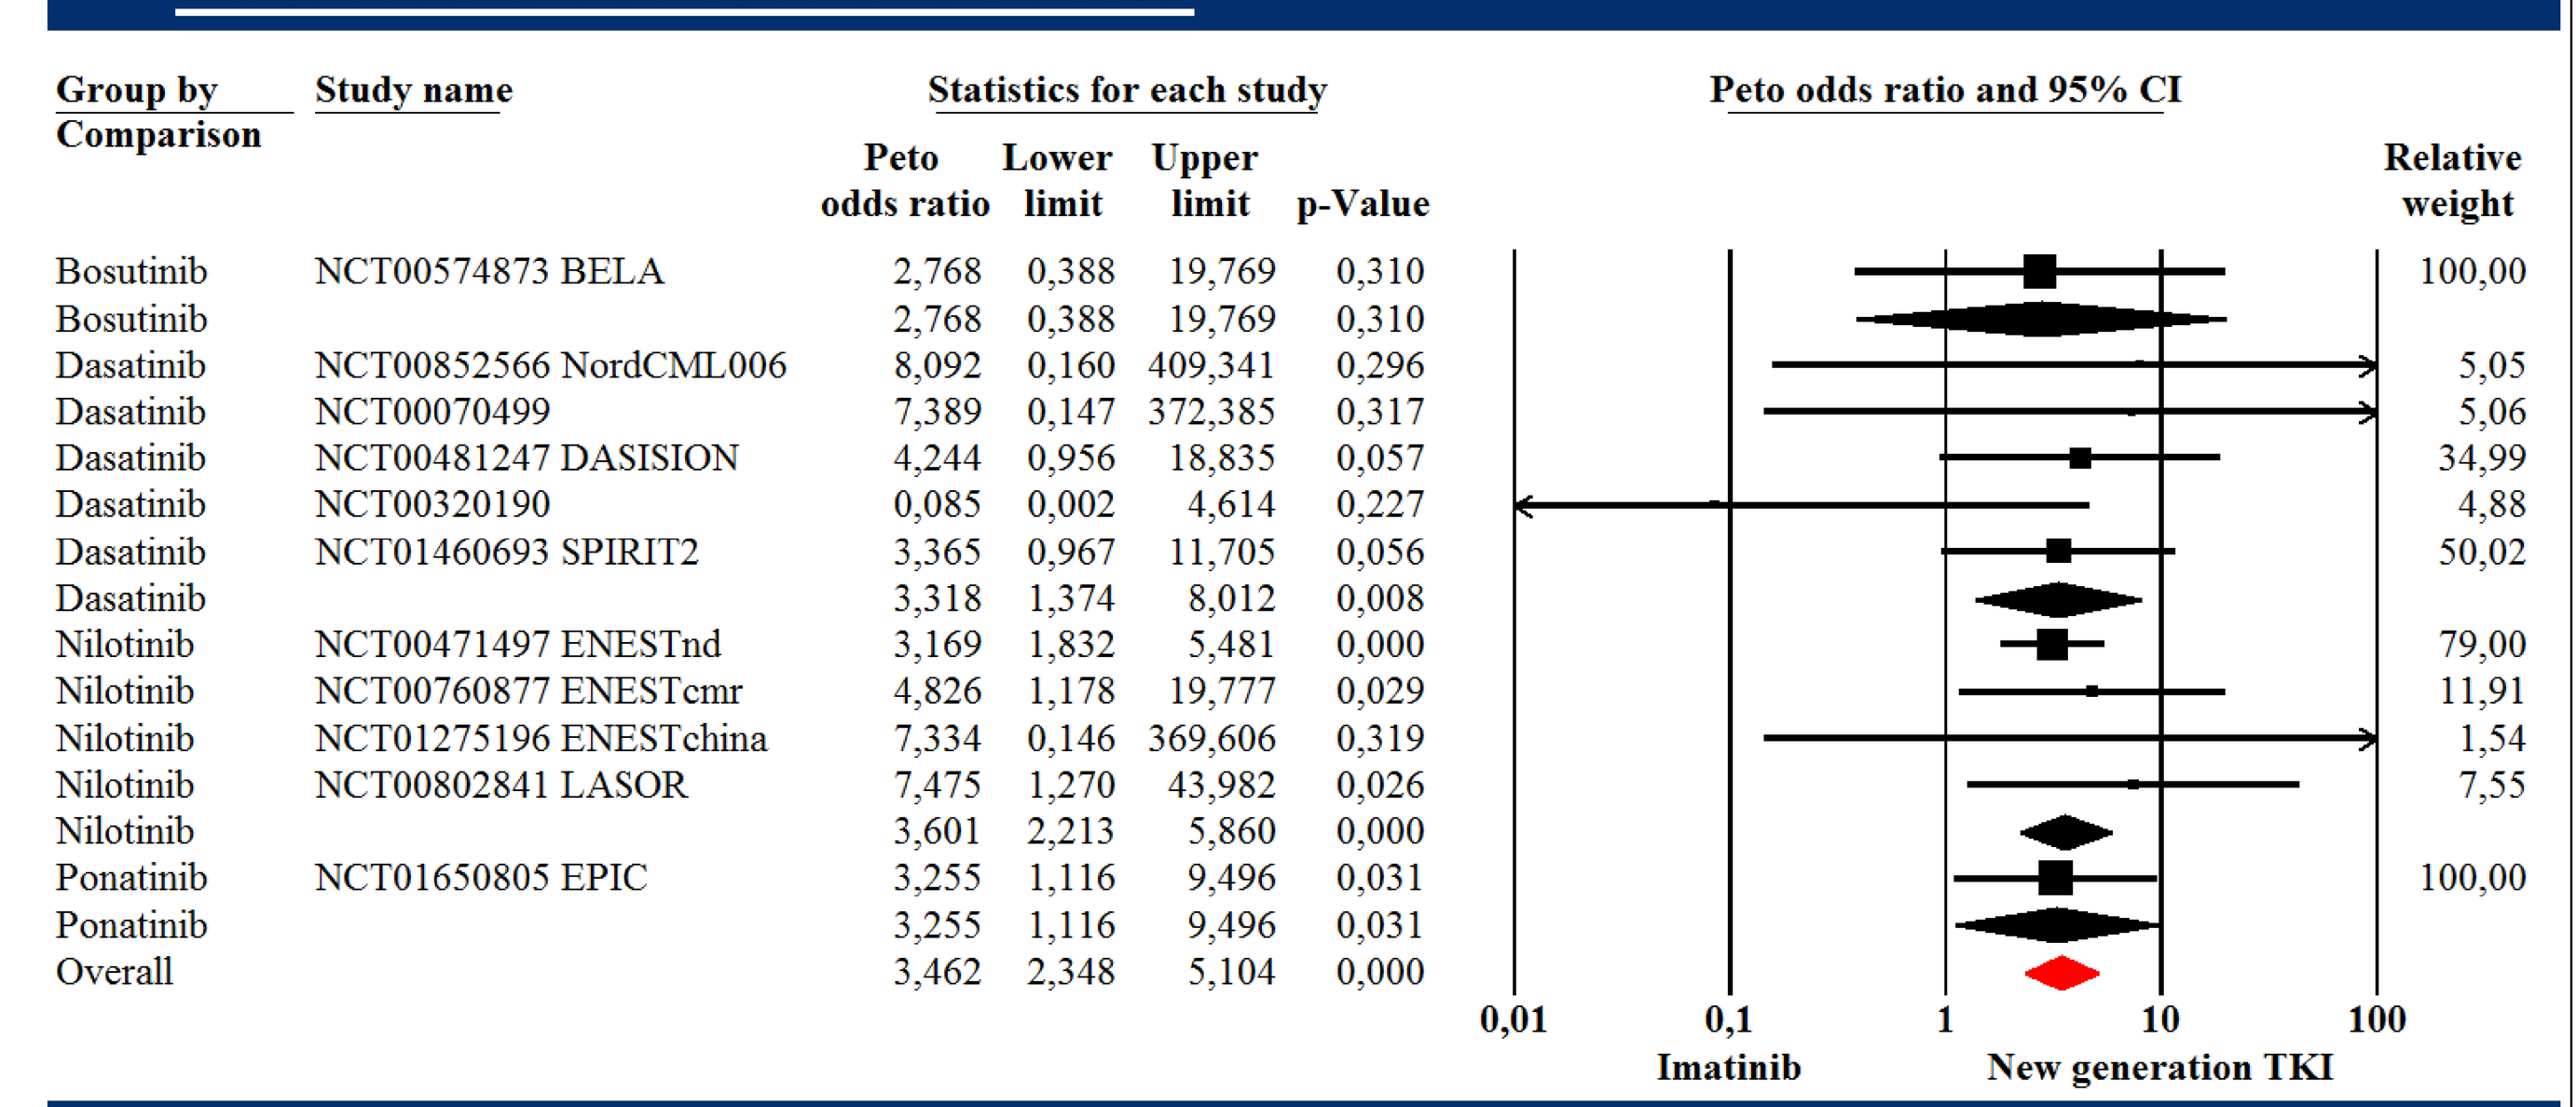
Group by Comparison
Study name
Statistics for each study
Peto odds ratio | Lower limit | Upper limit | p-Value
Peto odds ratio and 95% CI
Relative weight
Bosutinib NCT00574873 BELA 2,768 0,388 19,769 0,310 100,00
Bosutinib 2,768 0,388 19,769 0,310
Dasatinib NCT00852566 NordCML006 8,092 0,160 409,341 0,296 5,05
Dasatinib NCT00070499 7,389 0,147 372,385 0,317 5,06
Dasatinib NCT00481247 DASISION 4,244 0,956 18,835 0,057 34,99
Dasatinib NCT00320190 0,085 0,002 4,614 0,227 4,88
Dasatinib NCT01460693 SPIRIT2 3,365 0,967 11,705 0,056 50,02
Dasatinib 3,318 1,374 8,012 0,008
Nilotinib NCT00471497 ENESTnd 3,169 1,832 5,481 0,000 79,00
Nilotinib NCT00760877 ENESTcmr 4,826 1,178 19,777 0,029 11,91
Nilotinib NCT01275196 ENESTchina 7,334 0,146 369,606 0,319 1,54
Nilotinib NCT00802841 LASOR 7,475 1,270 43,982 0,026 7,55
Nilotinib 3,601 2,213 5,860 0,000
Ponatinib NCT01650805 EPIC 3,255 1,116 9,496 0,031 100,00
Ponatinib 3,255 1,116 9,496 0,031
Overall 3,462 2,348 5,104 0,000
0,01 0,1 1 10 100
Imatinib
New generation TKI

Venous occlusive events

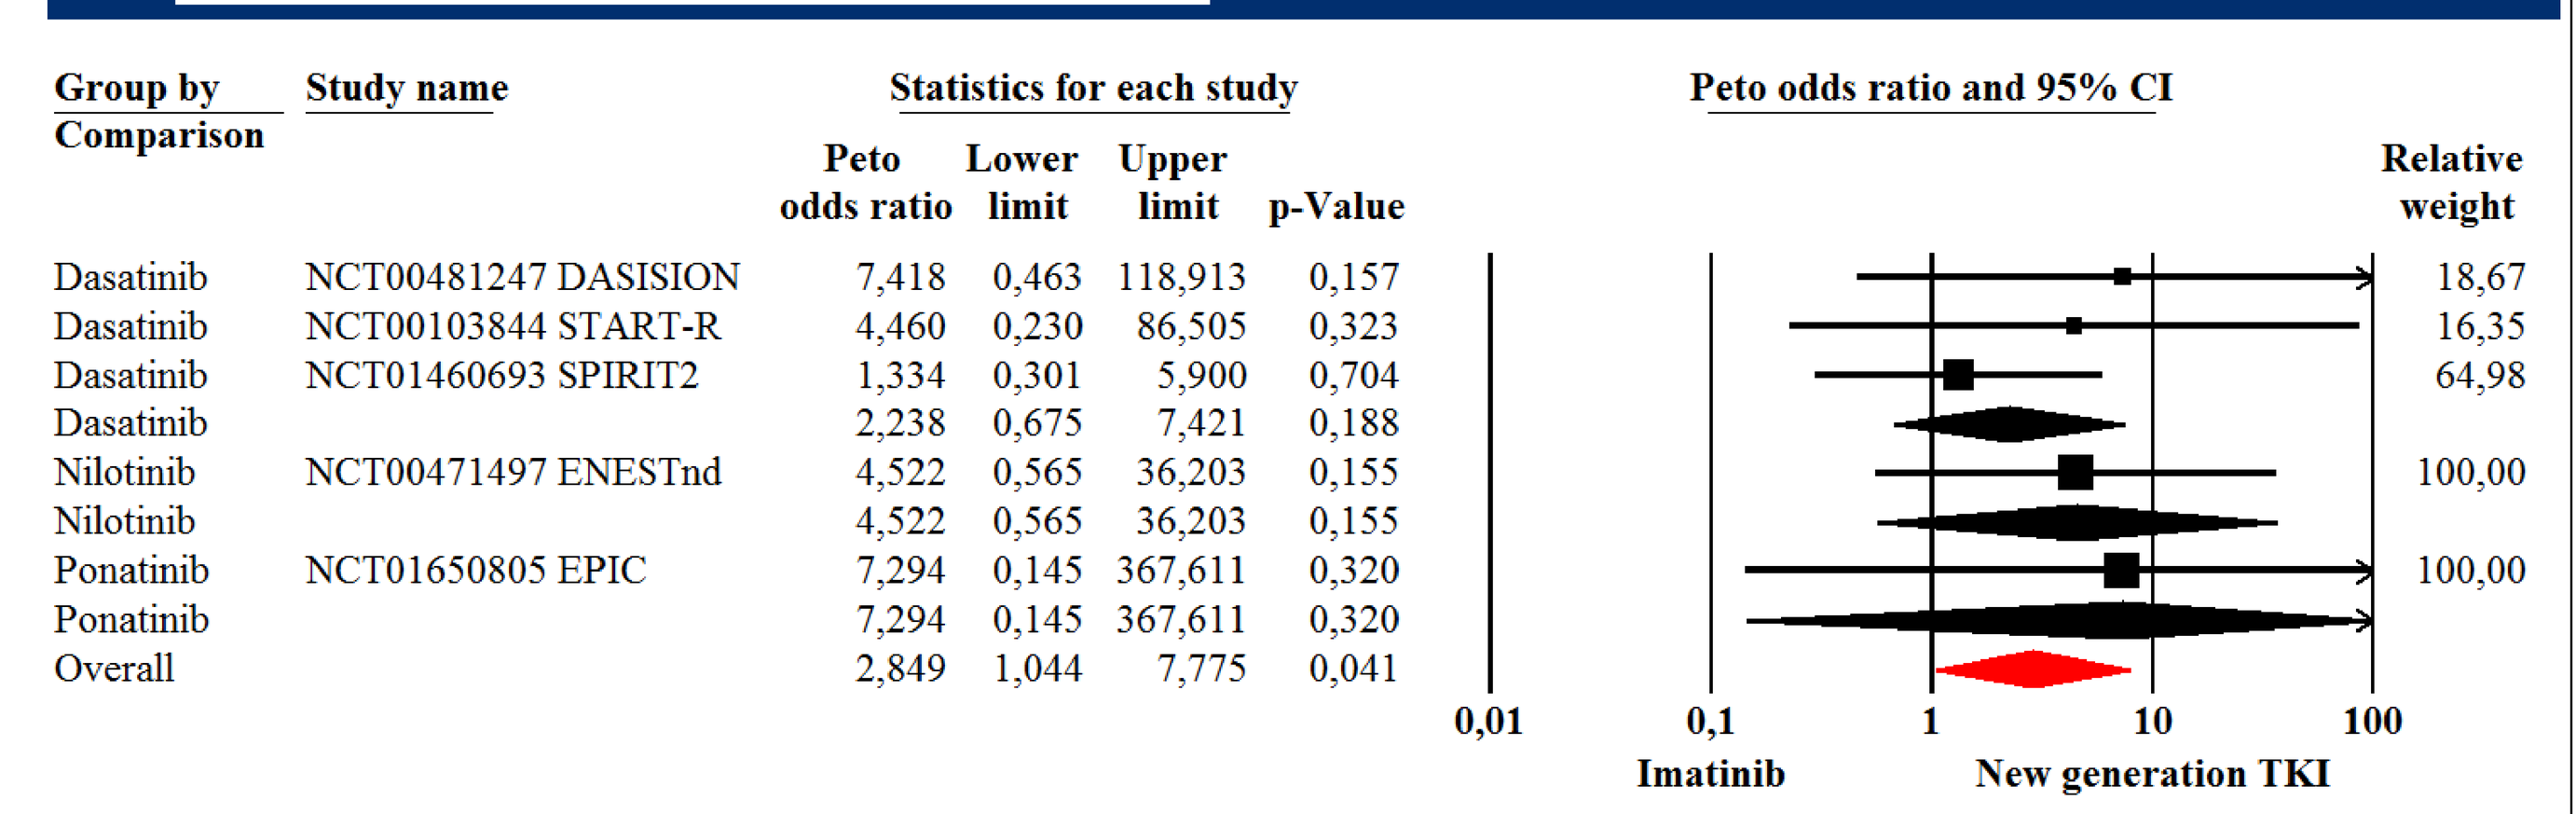
Group by Comparison
Study name
Statistics for each study
Peto odds ratio | Lower limit | Upper limit | p-Value
Peto odds ratio and 95% CI
Relative weight
Dasatinib NCT00481247 DASISION 7,418 0,463 118,913 0,157 18,67
Dasatinib NCT00103844 START-R 4,460 0,230 86,505 0,323 16,35
Dasatinib NCT01460693 SPIRIT2 1,334 0,301 5,900 0,704 64,98
Dasatinib 2,238 0,675 7,421 0,188
Nilotinib NCT00471497 ENESTnd 4,522 0,565 36,203 0,155 100,00
Nilotinib 4,522 0,565 36,203 0,155
Ponatinib NCT01650805 EPIC 7,294 0,145 367,611 0,320 100,00
Ponatinib 7,294 0,145 367,611 0,320
Overall 2,849 1,044 7,775 0,041
0,01 0,1 1 10 100
Imatinib
New generation TKI

- Ponatinib (REM OR_{PETO}: 3.26; 95%CI: 1.12 to 9.50), nilotinib (REM OR_{PETO}: 3.60; 95%CI: 2.21 to 5.86) and dasatinib (REM OR_{PETO}: 3.32; 95%CI: 1.37 to 8.01) are associated with higher risk of arterial occlusive events than imatinib.
- No significant difference was found with bosutinib but a trend indicate an increased risk of arterial occlusive events.
- Overall, new generation TKIs increase the rate of venous occlusive events (REM OR_{PETO}: 2.85; 95%CI: 1.04 to 7.78).
- Stratification by treatment for venous analysis demonstrates nonsignificant results due to the low power of the analysis.

Limitations

- Lack of time-to-event analyses
- Inconsistent report of cardiovascular events in the literature.
- However, the use of a clinical trial register aimed to decrease this heterogeneity, and funnel plots demonstrate no evidences of publication bias. The I^2 statistic specifies no heterogeneity among studies (data not shown).

Conclusions

- New generation TKIs increased risk of arterial and venous occlusive events compared with imatinib
- The increased risk of vascular occlusive events associated with new generation BCR-ABL TKIs is mainly driven by thrombotic events occurring at the arterial side.
- Additional investigations are required to assess the underlying pathophysiological mechanisms.
- Appropriate risk minimization measures should be taken/implemented with nilotinib, dasatinib and ponatinib.

Disclosures

François Mullier reports personal fees from Boehringer Ingelheim, Bayer Healthcare and Bristol-Myers Squibb-Pfizer outside the submitted work. Carlos Graux reports personal fees from Novartis, Celgene, and Amgen, outside the submitted work. The other authors have no conflict of interest to disclose.

Contact information

H. Haguet: helene.haguet@gmail.com
J. Douxfils: jonathan.douxfils@unamur.be
J-M Dogné: jean-michel.dogne@unamur.be

References

1. Giles FJ, Mauro MJ, Hong F, Ortmann CE, McNeill C, Woodman RC, et al. Rates of peripheral arterial occlusive disease in patients with chronic myeloid leukemia in the chronic phase treated with imatinib, nilotinib, or non-tyrosine kinase therapy: a retrospective cohort analysis. Leukemia. 2013;27(6):1310-5.
2. Quintás-Cardama A, Kantarjian H, Cortes J. Nilotinib-associated vascular events. Clinical lymphoma, myeloma & leukemia. 2012;12(5):337-40.
3. Douxfils J, Haguet H, Mullier F, Chatelain C, Graux C, Dogné JM. Association Between BCR-ABL Tyrosine Kinase Inhibitors for Chronic Myeloid Leukemia and Cardiovascular Events, Major Molecular Response, and Overall Survival: A Systematic Review and Meta-analysis. JAMA oncology. 2016.

MAY 25-28
MONTPELLIER